P1397

Verekitug, a thymic stromal lymphopoietin receptor antagonist, administered in participants with chronic rhinosinusitis with nasal polyps and comorbid asthma in the VIBRANT trial

Joseph K. Han,[1] Piotr H. Skarzynski,[2,3] Neetu Talreja,[4] Martha Tarpay,[5] Tanya M. Laidlaw,[6] José Miguel Villacampa Aubá,[7] Isam Alobid,[8–10] Joaquim Mullol,[8–10] Sven Becker,[11] Sumathi Sivapalasingam,[12] Kiran Patel,[12]* Ashish Kalra,[12] Arkadeep Sinha,[12] Richard Lin,[12] Fang Xie,[12] Andrzej Dymek,[13] Grazyna Pulka,[14] Aaron Deykin[12]

[1]Old Dominion University, Norfolk, VA, USA; [2]Center of Hearing and Speech MEDINCUS, Kajetany, Poland; [3]Institute of Sensory Organs, Kajetany, Poland; [4]The Allergy Group and Treasure Valley Medical Research, Boise, ID, USA; [5]Allergy, Asthma & Clinical Research Center, Oklahoma City, OK, USA; [6]Brigham and Women's Hospital, Harvard Medical School, Boston, MA, USA; [7]Hospital Universitario Fundación Jiménez Díaz, Madrid, Spain; [8]Hospital Clinic, Universitat de Barcelona, Barcelona, Catalonia, Spain; [9]IDIBAPS, Barcelona, Catalonia, Spain; [10]CIBERES, Barcelona, Catalonia, Spain; [11]University of Tübingen, Tübingen, Germany; [12]Upstream Bio, Inc., Waltham, MA, USA; [13]Centrum Medyczne Lucyna Andrzej Dymek, Strzelce Opolskie, Poland; [14]Centrum Medyczne ALL-MED-Badania Kliniczne, Kraków, Poland. *Affiliation at time of study.

SCAN ME
to download an electronic version of the poster and to access supplemental content. The QR code is intended to provide scientific information for individual reference. The PDF should not be altered or reproduced in any way.

KEY FINDINGS

Verekitug is a novel, high-affinity, fully human monoclonal antibody targeting the thymic stromal lymphopoietin receptor (TSLPR).

In the VIBRANT trial of participants with inadequately controlled chronic rhinosinusitis with nasal polyps (CRSwNP), approximately 60% had comorbid asthma with poor asthma symptom control as measured by the Asthma Control Questionnaire-6 (ACQ-6) but normal lung function as measured by forced expiratory volume in 1 second (FEV_1).

Verekitug 100 mg every 12 weeks (q12w) led to similar and significant improvements in nasal polyp score (NPS) in participants with and without comorbid asthma.

Verekitug led to generally greater reduction in sinonasal symptoms and Lund-Mackay (LMK) score in participants with comorbid asthma.

Need for rescue corticosteroids or sinus surgery was reduced in those with or without comorbid asthma.

Verekitug led to improved asthma symptom control by ACQ-6 but no change in FEV_1.

RATIONALE

- CRSwNP is characterized by high symptom burden and reduced quality of life, with thymic stromal lymphopoietin (TSLP) playing a key role in driving inflammation[1,2]
 - Up to 70% of patients with CRSwNP have comorbid asthma[3]
- Verekitug is a highly potent, fully human monoclonal antibody targeting the TSLPR[4]
- The phase 2 VIBRANT trial (NCT06164704) demonstrated that verekitug led to statistically and clinically significant improvements in NPS and sinonasal symptoms over 24 weeks in participants with severe, uncontrolled CRSwNP vs placebo[5]
 - The least squares mean (LSM) difference in NPS at 24 weeks for verekitug vs placebo was −1.77; $P < 0.0001$
- Here, we report the effect of verekitug on clinical endpoints, lung function, and asthma control in participants with comorbid asthma in the VIBRANT trial

METHODS

- VIBRANT, a double-blind, placebo-controlled, phase 2 trial, randomized participants with severe, uncontrolled CRSwNP (**Figure 1**)
- The treatment effect of verekitug was analyzed in participants with or without comorbid asthma via a mixed model for repeated measures using worst observed values to account for systemic corticosteroids (SCS) and worst possible values to account for rescue surgery

Figure 1. VIBRANT study design

Participant population
- Aged 18–85 years
- History of sinus surgery or NP exacerbation requiring SCS in the past 24 months, or contraindication/intolerance to SCS
- Endoscopic NPS ≥ 5
- NCS ≥ 2 over 14 days
- CRSwNP symptoms for ≥ 8 weeks
- Stable SoC treatment for CRSwNP for ≥ 30 days

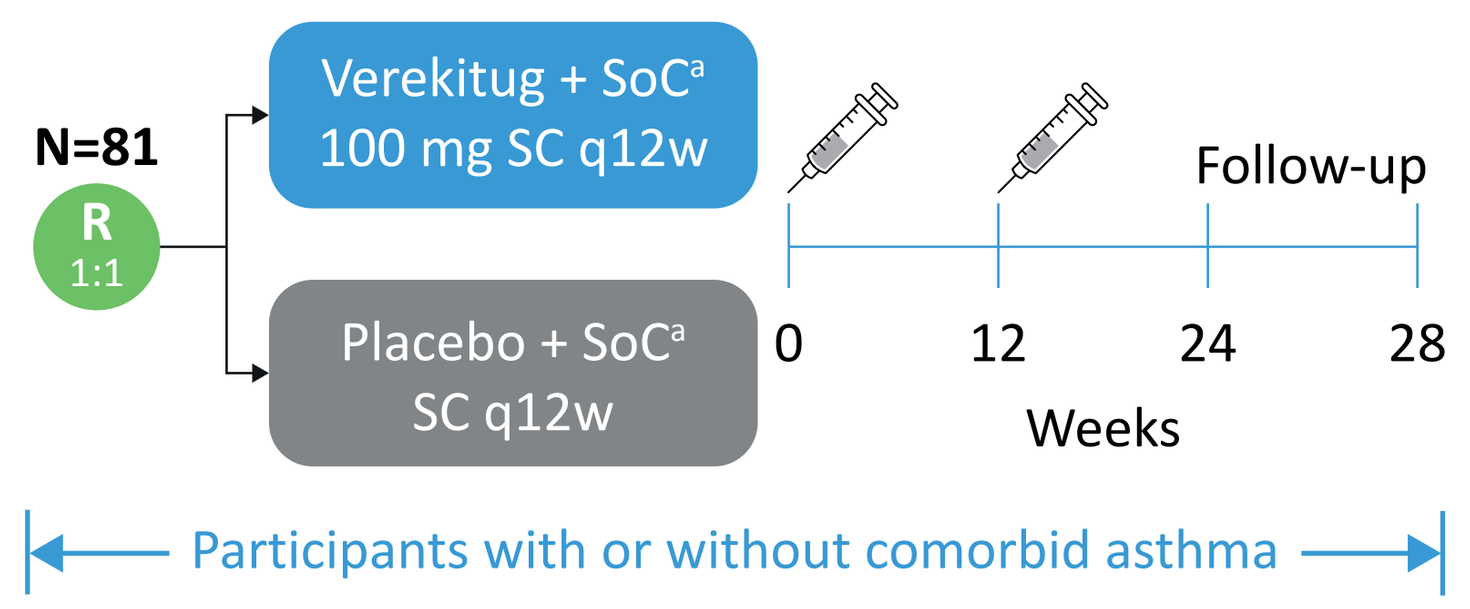

N=81
R
1:1
Verekitug + SoC[a]
100 mg SC q12w
Placebo + SoC[a]
SC q12w
Follow-up
0
12
24
28
Weeks
Participants with or without comorbid asthma

VIBRANT endpoints	
Primary endpoint	Change from baseline in endoscopic NPS at week 24
Key secondary endpoints	• Change from baseline at week 24 in – NCS – TSS – DSS – LMK score • Proportion requiring surgery/SCS by week 24
Other endpoints[b]	• Change from baseline at week 24 in – ACQ-6 – Pre-bronchodilator FEV_1
Safety	TEAEs

Syringes indicate first and second dose.
[a]Intranasal corticosteroid. [b]Assessed in participants with comorbid asthma only.
DSS, difficulty with sense of smell; NCS, nasal congestion score; SC, subcutaneous; SoC, standard of care; TEAE, treatment-emergent adverse event; TSS, total symptom score.

RESULTS

- 58% (47/81) of participants had comorbid asthma with or without nonsteroidal anti-inflammatory drug-exacerbated respiratory disease (NSAID-ERD)
- 28% (13/47) of participants had both comorbid asthma and NSAID-ERD; no participant had a diagnosis of NSAID-ERD only
- Participants with comorbid asthma had poor asthma control per ACQ-6 but normal lung function per FEV_1 (**Table 1**)
- 72% (33/46) of participants with comorbid asthma had blood eosinophils of ≥ 300/µL (**Supplementary Table 1**, accessible via the QR code)

RESULTS

Table 1. Balanced baseline characteristics across treatment arms

	Comorbid asthma		No asthma	
	Verekitug (n=24)	**Placebo (n=23)**	**Verekitug (n=17)**	**Placebo (n=17)**
Median age (range), years	47 (19–75)	49 (21–66)	53 (30–74)	52 (28–76)
Female sex, n (%)	14 (58.3)	9 (39.1)	1 (5.9)	5 (29.4)
NPS (0–8), mean (SD)	5.9 (1.4)	6.0 (1.4)	5.9 (2.0)	6.3 (1.2)
NCS (0–3), mean (SD)	2.6 (0.4)	2.7 (0.5)	2.6 (0.4)	2.7 (0.4)
TSS (0–24), mean (SD)	18.1 (4.2)	18.3 (5.6)	16.8 (4.6)	18.1 (4.6)
DSS (0–3), mean (SD)	2.9 (0.3)	2.9 (0.3)	2.8 (0.4)	2.6 (0.8)
LMK score (0–24), mean (SD)	18.3 (3.8)	18.1 (4.9)[a]	15.2 (5.7)[b]	15.5 (4.7)
SCS in past 24 months, n (%)	8 (33.3)	10 (43.5)	11 (64.7)	9 (52.9)
Sinus surgery, n (%)	18 (75.0)	18 (78.3)	8 (47.1)	8 (47.1)
ACQ-6, mean (SD)	1.5 (1.1)[c]	2.4 (1.3)[d]	–	–
ACQ-6 ≥ 1.5, n (%)	10 (41.7)	15 (65.2)	–	–
Mean prebronchodilator FEV_1 predicted (SD), %	86.5 (13.7)	85.6 (19.6)[e]	–	–
FEV_1 < 85%, n (%)	10 (41.7)	9 (39.1)	–	–

[a]n=19. [b]n=15. [c]n=22. [d]n=19. [e]n=22.
SD, standard deviation; TSS, total symptom score.

- Verekitug improved **NPS** at 24 weeks compared with placebo in participants with CRSwNP with or without comorbid asthma similarly (**Figure 2**), and consistent with the overall VIBRANT population[5]

Figure 2. Verekitug treatment effect on NPS at week 24 in participants with or without comorbid asthma

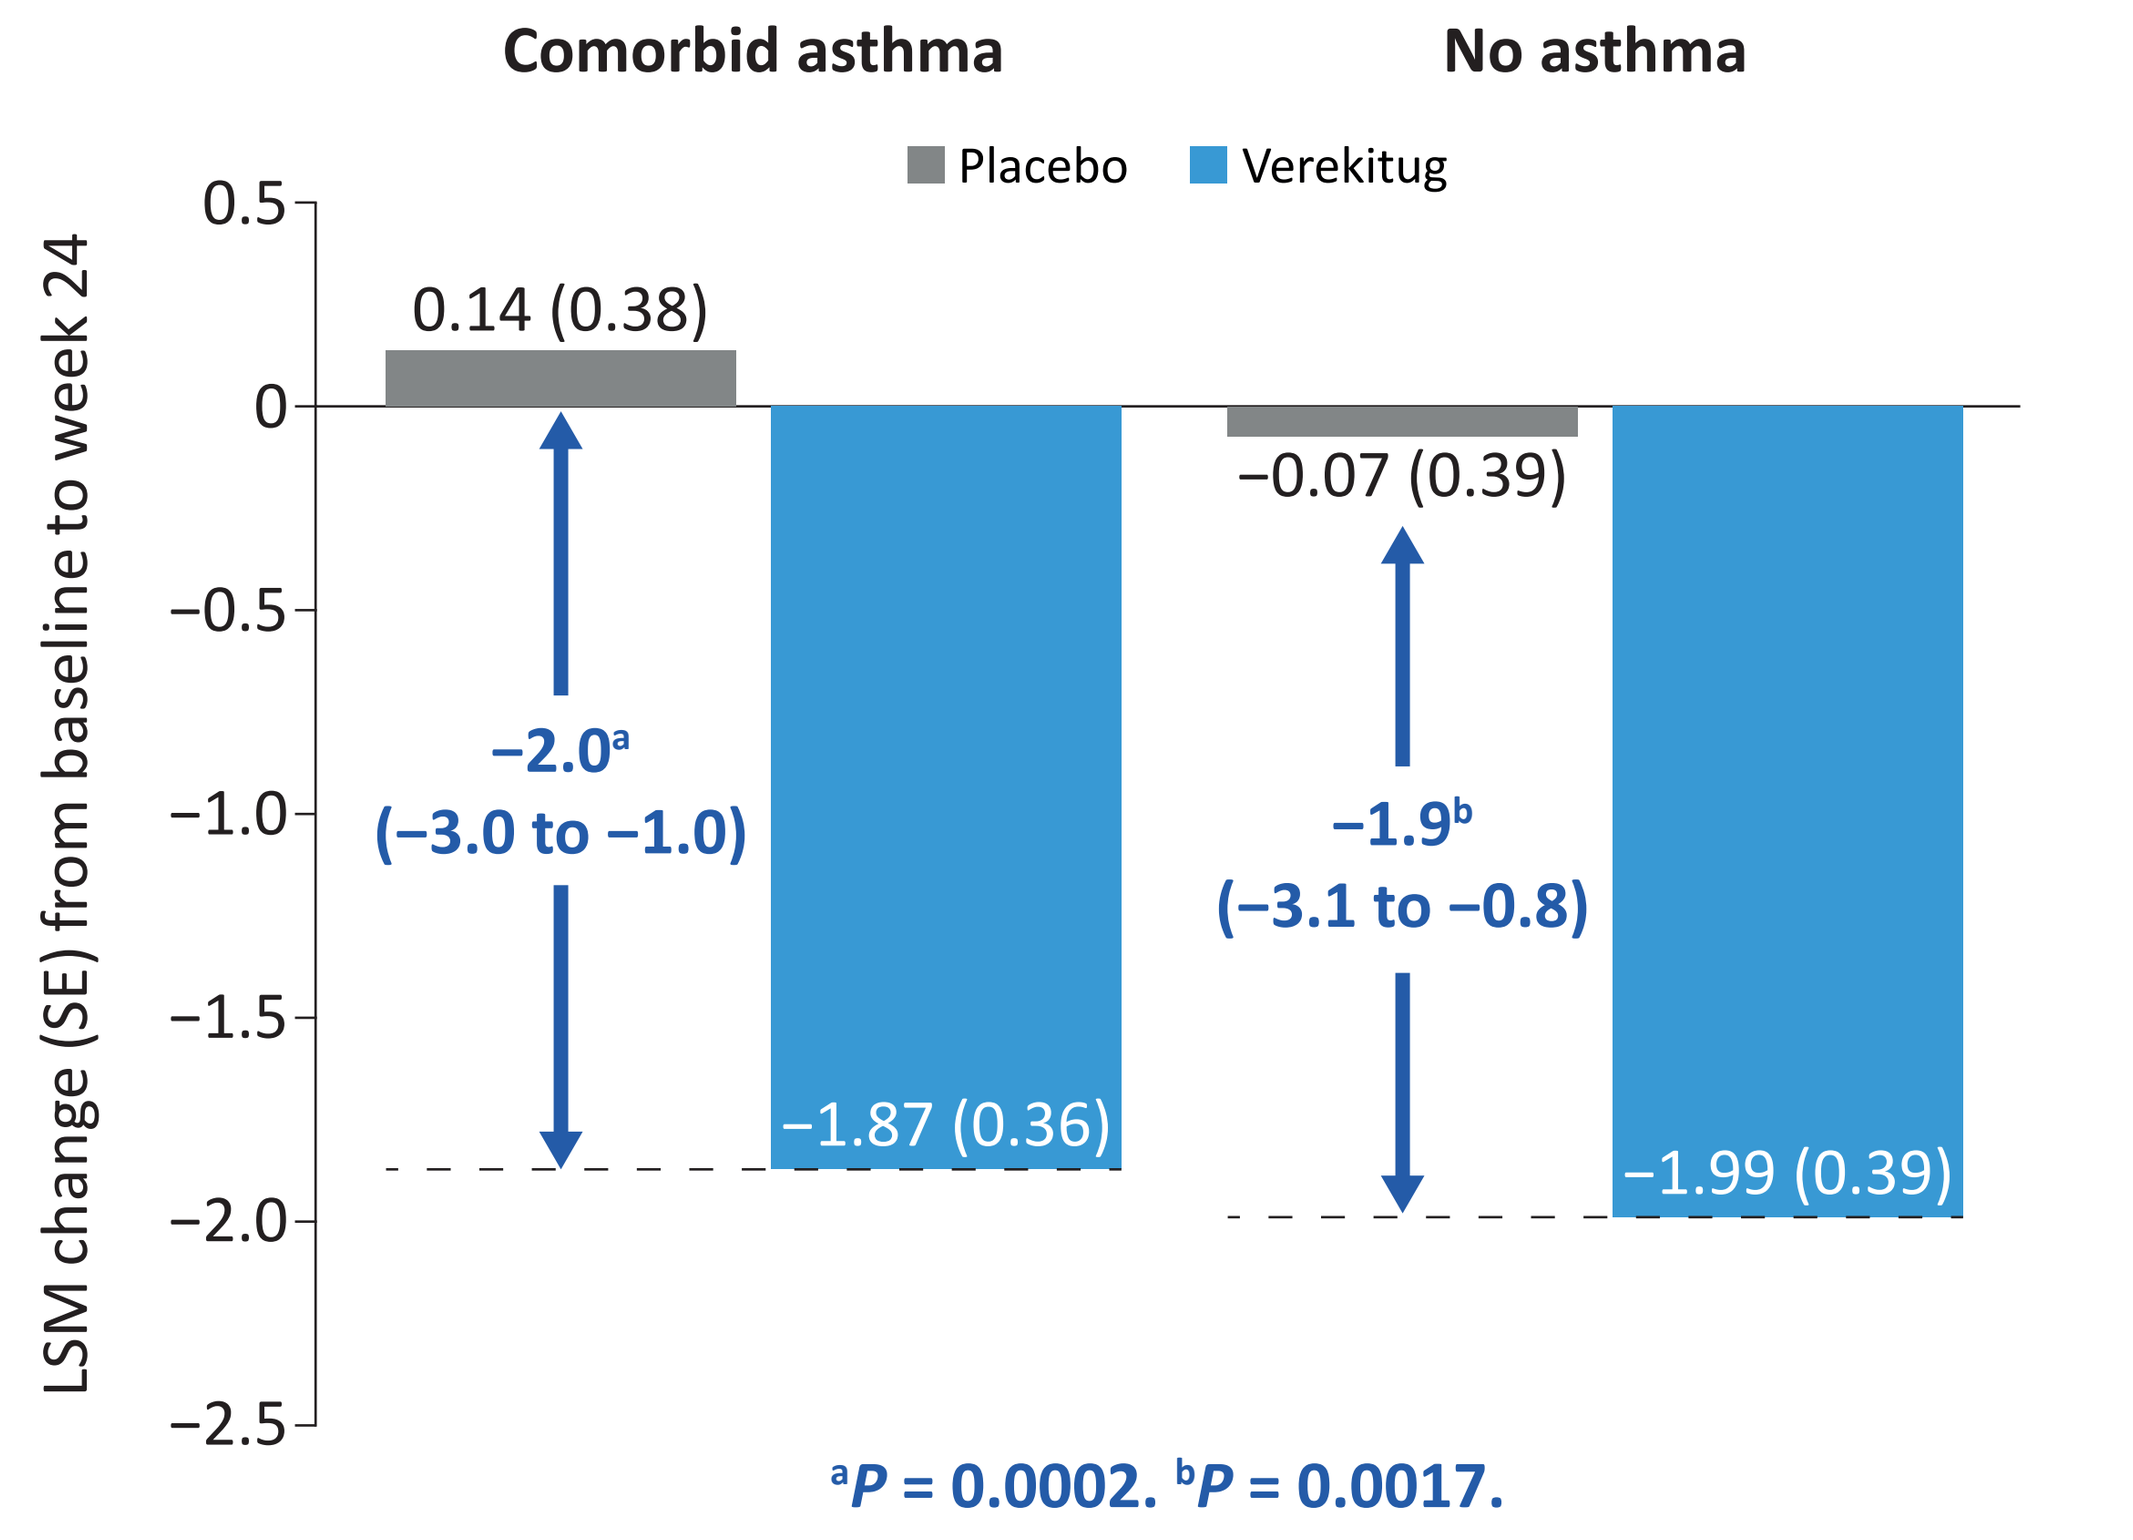

Comorbid asthma
No asthma
Placebo
Verekitug
LSM change (SE) from baseline to week 24
0.5
0
−0.5
−1.0
−1.5
−2.0
−2.5
0.14 (0.38)
−2.0[a]
(−3.0 to −1.0)
−1.87 (0.36)
−0.07 (0.39)
−1.9[b]
(−3.1 to −0.8)
−1.99 (0.39)
[a]P = 0.0002. [b]P = 0.0017.

LSM differences (95% CI) and *P* values for verekitug vs placebo are shown in dark blue.
SE, standard error.

- Verekitug improved **sinonasal symptoms** and **LMK score** compared with placebo regardless of comorbid asthma (**Figure 3**)
- Generally greater improvements in **NCS**, **TSS**, **DSS** (loss of smell), and **LMK score** were observed in participants with comorbid asthma than without (**Figure 3**)
- Verekitug improved asthma symptom control measured by **ACQ-6** compared with placebo (**Figure 3**)
- No treatment effect was observed for FEV_1 (**Figure 3**), likely due to generally normal lung function at baseline

Figure 3. Verekitug treatment effect on outcomes at week 24 in participants with or without comorbid asthma

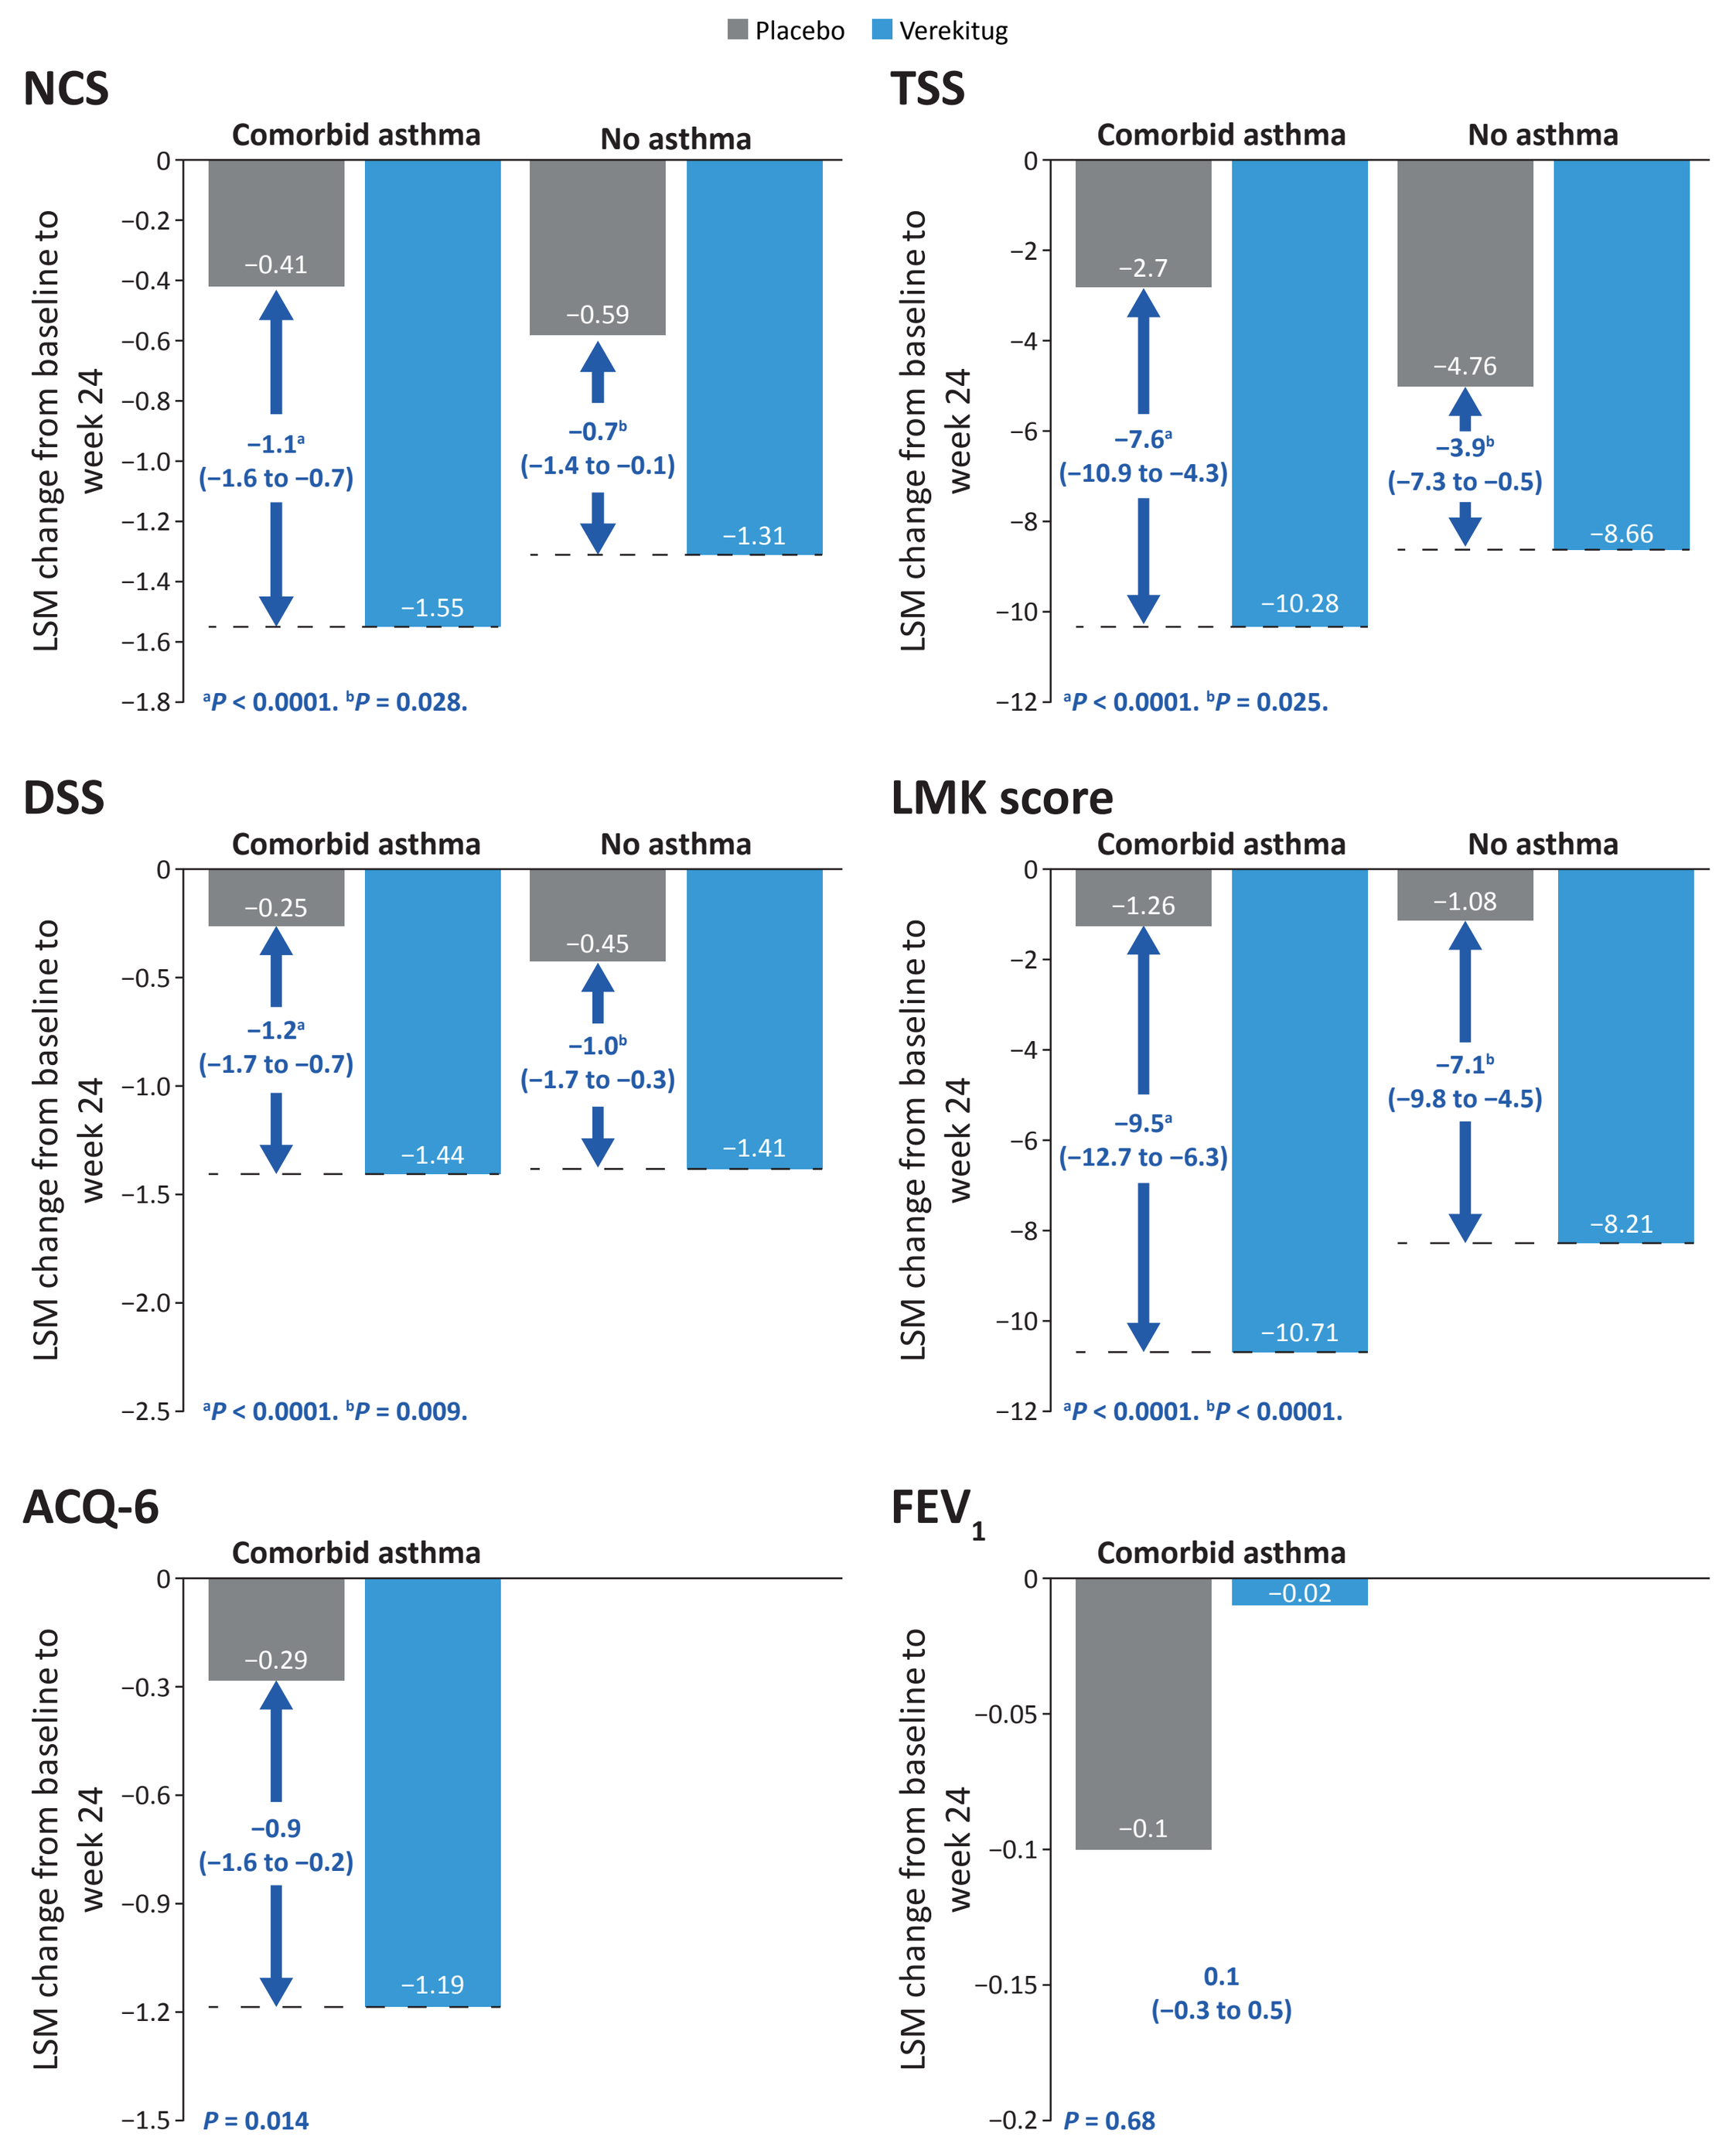

Placebo
Verekitug
NCS
Comorbid asthma
No asthma
LSM change from baseline to week 24
−0.41
−1.1[a]
(−1.6 to −0.7)
−1.55
−0.59
−0.7[b]
(−1.4 to −0.1)
−1.31
[a]P < 0.0001. [b]P = 0.028.
TSS
−2.7
−7.6[a]
(−10.9 to −4.3)
−10.28
−4.76
−3.9[b]
(−7.3 to −0.5)
−8.66
[a]P < 0.0001. [b]P = 0.025.
DSS
−0.25
−1.2[a]
(−1.7 to −0.7)
−1.44
−0.45
−1.0[b]
(−1.7 to −0.3)
−1.41
[a]P < 0.0001. [b]P = 0.009.
LMK score
−1.26
−9.5[a]
(−12.7 to −6.3)
−10.71
−1.08
−7.1[b]
(−9.8 to −4.5)
−8.21
[a]P < 0.0001. [b]P < 0.0001.
ACQ-6
−0.29
−0.9
(−1.6 to −0.2)
−1.19
P = 0.014
FEV1
−0.1
−0.02
0.1
(−0.3 to 0.5)
P = 0.68

LSM differences (95% CI) and *P* values for verekitug vs placebo are shown in dark blue.

- There was an 83% reduction in need for **rescue SCS or sinus surgery** in participants with comorbid asthma (nominal $P = 0.027$) and a 53% reduction in participants without comorbid asthma (nominal $P = 0.55$)

- Verekitug was generally well tolerated by participants with or without comorbid asthma (**Table 2**), with the safety profile being consistent with the overall population[5]

Table 2. TEAEs[a] in participants with or without comorbid asthma

	Comorbid asthma		No asthma	
TEAEs, n (%)	**Verekitug (n=23)**	**Placebo (n=23)**	**Verekitug (n=17)**	**Placebo (n=17)**
All TEAEs	14 (60.9)	15 (65.2)	13 (76.5)	11 (64.7)
Related to study treatment[a]	1 (4.3)	3 (13.0)	0	0
Serious	0	0	0	0
Grade ≥ 3	1 (4.3)	5 (21.7)	1 (5.9)	0
Leading to study drug discontinuation	0	1 (4.3)	0	0

[a]Adverse events that were assessed by the investigator as related, or adverse events with missing relationship to study treatment, were assumed to be related to study treatment.

CONCLUSIONS

- In the VIBRANT trial, ≈ 60% of participants with inadequately controlled CRSwNP had comorbid asthma with evidence of poor control of asthma symptoms by ACQ-6 but normal lung function by FEV_1
- Verekitug treatment improved NPS at week 24 in those with and without comorbid asthma, and was consistent with the overall VIBRANT population
- Verekitug led to generally greater improvement in CRSwNP symptoms and LMK score in those with comorbid asthma vs those without
- A greater reduction in the need for rescue corticosteroids or sinus surgery was seen in those with comorbid asthma
- Verekitug led to improvements in ACQ-6 scores with no impact on FEV_1

REFERENCES

1. Fokkens WJ, et al. *Rhinology*. 2020;58(suppl 29):1–464. **2.** Corren J, Ziegler SF. *Nat Immunol*. 2019;20:1603–1609. **3.** Bachert C, et al. *J Allergy Clin Immunol Pract*. 2023;11:2630–2641. **4.** Kalra A, et al. *Eur Respir J*. 2025;66(suppl 69):OA5392. **5.** Han JK, et al. *J Allergy Clin Immunol*. 2026;157(suppl 2):AB432.

ACKNOWLEDGMENTS

This study was funded by Upstream Bio, Inc., Waltham, MA, USA. Writing assistance was provided by Larissa Belova, PhD, of Parexel, and was funded by Upstream Bio, Inc.

Presented at the 2026 American Thoracic Society (ATS) International Conference; May 15–20; Orlando, FL, USA

P1397

Verekitug, a thymic stromal lymphopoietin receptor antagonist, administered in participants with chronic rhinosinusitis with nasal polyps and comorbid asthma in the VIBRANT trial

Joseph K. Han,[1] Piotr H. Skarzynski,[2,3] Neetu Talreja,[4] Martha Tarpay,[5] Tanya M. Laidlaw,[6] José Miguel Villacampa Aubá,[7] Isam Alobid,[8–10] Joaquim Mullol,[8–10] Sven Becker,[11] Sumathi Sivapalasingam,[12] Kiran Patel,[12]* Ashish Kalra,[12] Arkadeep Sinha,[12] Richard Lin,[12] Fang Xie,[12] Andrzej Dymek,[13] Grazyna Pulka,[14] Aaron Deykin[12]

[1]Old Dominion University, Norfolk, VA, USA; [2]Center of Hearing and Speech MEDINCUS, Kajetany, Poland; [3]Institute of Sensory Organs, Kajetany, Poland; [4]The Allergy Group and Treasure Valley Medical Research, Boise, ID, USA; [5]Allergy, Asthma & Clinical Research Center, Oklahoma City, OK, USA; [6]Brigham and Women's Hospital, Harvard Medical School, Boston, MA, USA; [7]Hospital Universitario Fundación Jiménez Díaz, Madrid, Spain; [8]Hospital Clinic, Universitat de Barcelona, Barcelona, Catalonia, Spain; [9]IDIBAPS, Barcelona, Catalonia, Spain; [10]CIBERES, Barcelona, Catalonia, Spain; [11]University of Tübingen, Tübingen, Germany; [12]Upstream Bio, Inc., Waltham, MA, USA; [13]Centrum Medyczne Lucyna Andrzej Dymek, Strzelce Opolskie, Poland; [14]Centrum Medyczne ALL-MED-Badania Kliniczne, Kraków, Poland. *Affiliation at time of study.

SUPPLEMENTARY MATERIAL

Supplementary Table 1. Baseline characteristics of participants with or without comorbid asthma and by eosinophil levels

	Comorbid asthma (n=46)		No asthma (n=34)		All participants (N=80)	
	EOS < 300 cells/µL (n=13)	EOS ≥ 300 cells/µL (n=33)	EOS < 300 cells/µL (n=15)	EOS ≥ 300 cells/µL (n=19)	EOS < 300 cells/µL (n=28)	EOS ≥ 300 cells/µL (n=52)
Median age (range), years	52 (30–75)	45 (19–64)	52 (30–76)	53 (28–73)	52 (30–76)	49 (19–73)
Female sex, n (%)	4 (30.8)	19 (57.6)	3 (20.0)	3 (15.8)	7 (25.0)	22 (42.3)
NPS (0–8), mean (SD)	5.7 (1.0)	6.0 (1.5)	6.1 (1.4)	6.1 (1.8)	5.9 (1.2)	6.0 (1.6)
NCS (0–3), mean (SD)	2.7 (0.5)	2.6 (0.4)	2.8 (0.3)	2.5 (0.4)	2.7 (0.4)	2.6 (0.4)
TSS (0–24), mean (SD)	17.6 (5.7)	18.4 (4.6)	18.7 (4.2)	16.5 (4.8)	18.2 (4.9)	17.7 (4.7)
DSS (0–3), mean (SD)	3.0 (0.2)	2.8 (0.3)	2.7 (0.8)	2.7 (0.4)	2.8 (0.6)	2.8 (0.4)
LMK score (0–24), mean (SD)	18.8 (4.0)[a]	17.8 (4.4)[b]	15.1 (4.9)[c]	15.6 (5.4)[d]	16.8 (4.8)[e]	17.0 (4.9)[f]
SCS in past 24 months, n (%)	2 (15.4)	15 (45.5)	6 (40.0)	14 (73.7)	8 (28.6)	29 (55.8)
Sinus surgery, n (%)	10 (76.9)	25 (75.8)	9 (60.0)	7 (36.8)	19 (67.9)	32 (61.5)
ACQ-6, mean (SD)	2.3 (1.1)[g]	1.8 (1.3)[h]	-	-	-	-
ACQ-6 ≥ 1.5, n (%)	9 (69.2)	15 (45.5)	-	-	-	-
Mean prebronchodilator FEV_1 predicted (SD), %	87.2 (19.5)	85.3 (15.7)[i]	-	-	-	-
FEV_1 < 85%, n (%)	3 (23.1)	16 (48.5)	-	-	-	-

[a]n=12. [b]n=30. [c]n=14. [d]n=18. [e]n=26. [f]n=48. [g]n=12. [h]n=28. [i]n=32.
ACQ-6, Asthma Control Questionnaire-6; DSS, difficulty with sense of smell; EOS, eosinophils; FEV_1, forced expiratory volume in 1 second; LMK, Lund-Mackay; NP, nasal polyp; NCS, nasal congestion score; NPS, nasal polyp score; SCS, systemic corticosteroids; SD, standard deviation; TSS, total symptom score.

- Participants with comorbid asthma and eosinophils ≥ 300 cells/µL were younger, more often female, with higher SCS use and FEV_1 < 85% rates, and lower ACQ-6 scores versus those with < 300 cells/µL

Presented at the 2026 American Thoracic Society (ATS) International Conference; May 15–20; Orlando, FL, USA