P1398

Verekitug, a thymic stromal lymphopoietin receptor antagonist, in chronic rhinosinusitis with nasal polyps: effect on type 2 inflammatory biomarkers in the VIBRANT trial

SCAN ME
to download an electronic version of the poster and to access supplemental content. The QR code is intended to provide scientific information for individual reference. The PDF should not be altered or reproduced in any way.

Joseph K. Han,[1] Piotr H. Skarzynski,[2,3] Neetu Talreja,[4] Martha Tarpay,[5] Tanya M. Laidlaw,[6] José Miguel Villacampa Aubá,[7] Isam Alobid,[8–10] Joaquim Mullol,[8–10] Sven Becker,[11] Sumathi Sivapalasingam,[12] Kiran Patel,[12]* Ashish Kalra,[12] Arkadeep Sinha,[12] Fatema Chowdhury,[12] Richard Lin,[12] Fang Xie,[12] Andrzej Dymek,[13] Grazyna Pulka,[14] Aaron Deykin[12]

[1]Old Dominion University, Norfolk, VA, USA; [2]Center of Hearing and Speech MEDINCUS, Kajetany, Poland; [3]Institute of Sensory Organs, Kajetany, Poland; [4]The Allergy Group and Treasure Valley Medical Research, Boise, ID, USA; [5]Allergy, Asthma & Clinical Research Center, Oklahoma City, OK, USA; [6]Brigham and Women's Hospital, Harvard Medical School, Boston, MA, USA; [7]Hospital Universitario Fundación Jiménez Díaz, Madrid, Spain; [8]Hospital Clinic, Universitat de Barcelona, Barcelona, Catalonia, Spain; [9]IDIBAPS, Barcelona, Catalonia, Spain; [10]CIBERES, Barcelona, Catalonia, Spain; [11]University of Tübingen, Tübingen, Germany; [12]Upstream Bio, Inc., Waltham, MA, USA; [13]Centrum Medyczne Lucyna Andrzej Dymek, Strzelce Opolskie, Poland; [14]Centrum Medyczne ALL-MED-Badania Kliniczne, Kraków, Poland. *Affiliation at time of study.

KEY FINDINGS

Verekitug, a novel, high-affinity, fully human monoclonal antibody targeting the thymic stromal lymphopoietin receptor (TSLPR), when dosed every 12 weeks (q12w) in participants with chronic rhinosinusitis with nasal polyps (CRSwNP), achieved rapid and sustained suppression of type 2 inflammatory chemokines, cytokines, and biomarkers in both blood and nasal secretions, which may contribute to reduced nasal polyp score (NPS) and symptom burden.

RATIONALE

- CRSwNP is characterized by systemic and local inflammation, nasal polyp (NP) formation, and sinonasal symptoms[1–3]
- TSLPR signaling has emerged as a key driver of immune activation underlying CRSwNP morbidity[2]
- In the phase 2 VIBRANT trial (NCT06164704), treatment with verekitug, a TSLPR antagonist, led to statistically significant improvements vs placebo in NPS and nasal congestion score (NCS) over 24 weeks of treatment in participants with severe, uncontrolled CRSwNP[4,5]
- Here, we present a post hoc analysis of the effect of verekitug on nasal and blood inflammatory biomarker levels over 24 weeks in VIBRANT trial participants

REFERENCES

1. Yang C, et al. *Front Allergy*. 2025;6:1599797. **2.** Farokhi S, et al. *Immuno*. 2025;5:26. **3.** Del Toro E, et al. *StatPearls*. Updated May 5, 2025. Accessed March 5, 2026. https://www.ncbi.nlm.nih.gov/books/NBK560746. **4.** Han J, et al. Poster presented at the 2026 American Academy of Allergy, Asthma and Immunology (AAAAI) Annual Meeting; February 27–March 2; Philadelphia, PA, USA. Poster L60. **5.** ClinicalTrials.gov: NCT06164704. Updated August 15, 2025. Accessed January 23, 2026. https://clinicaltrials.gov/study/NCT06164704. **6.** Ito T, et al. *J Exp Med*. 2005;202:1213–1223. **7.** Blanchard C, Rothenberg ME. *Adv Immunol*. 2009;101:81–121. **8.** Izuhara K, et al. *Am J Respir Crit Care Med*. 2016;193:949–956.

METHODS

- VIBRANT is a double-blind, randomized phase 2 trial assessing the efficacy and safety of either 100 mg subcutaneous (SC) verekitug or matching placebo q12w, evaluated at 24 weeks, with standard of care (SoC) treatment (**Figure 1**)[5]
 - This post hoc analysis evaluated the change from baseline (CFB) in levels of inflammatory biomarkers in blood and nasal secretions over 24 weeks of verekitug treatment

Figure 1. VIBRANT study design

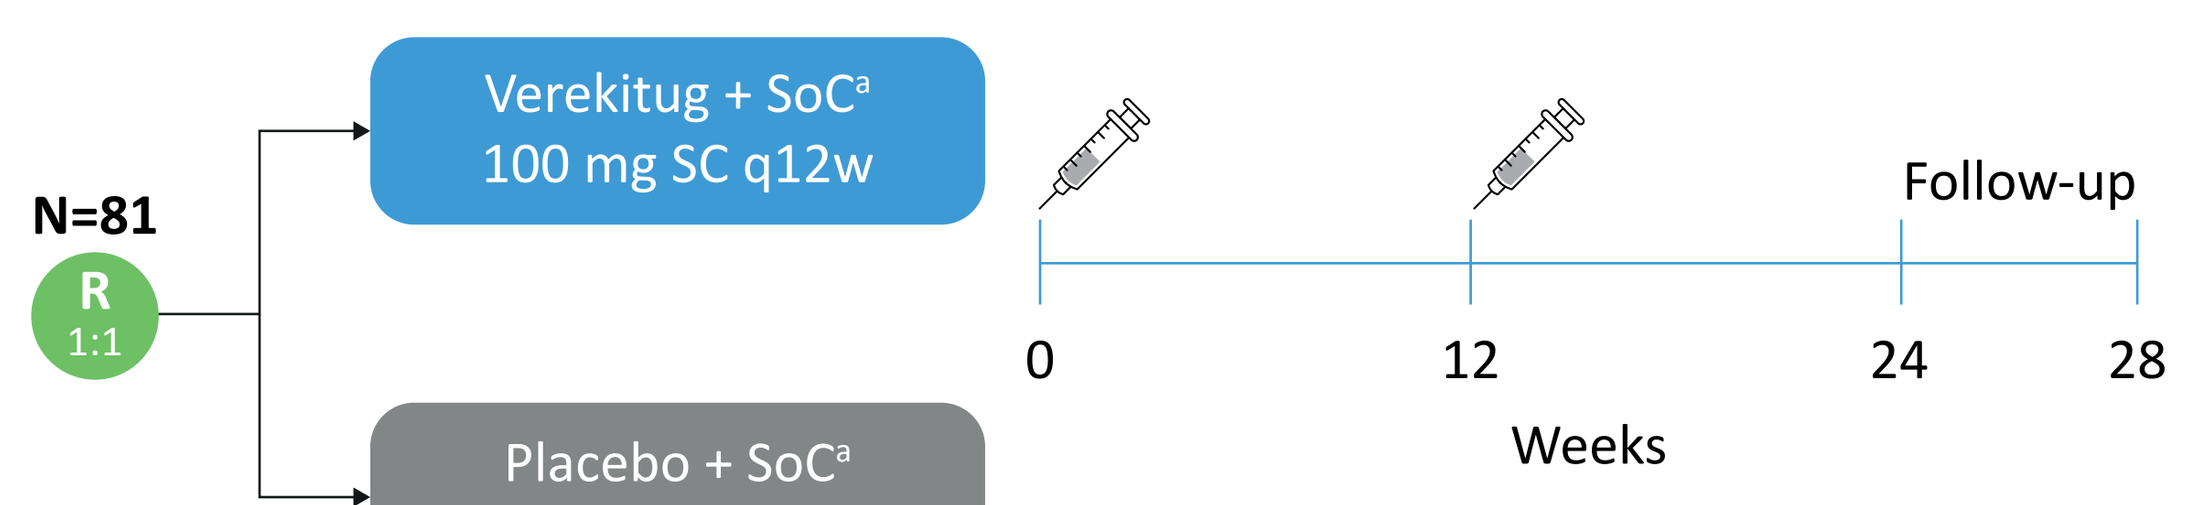

N=81
R
1:1
Verekitug + SoC[a]
100 mg SC q12w
Placebo + SoC[a]
SC q12w
Follow-up
0
12
24
28
Weeks

Participant population
- Aged 18–85 years
- History of sinus surgery or NP exacerbation requiring SCS in the past 24 months, or contraindication/intolerance to SCS
- Endoscopic NPS ≥ 5
- NCS ≥ 2 over 14 days
- CRSwNP symptoms for ≥ 8 weeks
- Stable SoC treatment for CRSwNP for ≥ 30 days

Syringes indicate first and second dose.
[a]Intranasal corticosteroid.
SCS, systemic corticosteroids.

RESULTS

- A total of 81 participants were randomized (41 to verekitug, 40 to placebo); baseline characteristics were balanced[4]

- Baseline levels of blood biomarkers were generally balanced across treatment groups, while some imbalances were seen in baseline levels of nasal biomarkers (**Supplementary Table 1**, accessible via the QR code)

ACKNOWLEDGMENTS

This study was funded by Upstream Bio, Inc., Waltham, MA, USA. Correlation analysis was performed by Leslie Abbott. Writing assistance was provided by Nivedita Saxena, PhD, of Parexel, and was funded by Upstream Bio, Inc.

RESULTS

- Verekitug reduced serum and nasal levels of type 2 cytokines: **IL-4**, **IL-5**, and **IL-13** (**Figure 2**)
- Greater reductions were seen in nasal secretions vs blood in **periostin**, **TARC**, **MDC**, and **eotaxin-3**—mediators known to contribute to local (nasal) inflammation and fibrotic activity[1,6–8] (**Figure 2**)

Figure 2. Percent change in type 2 biomarkers in nasal secretions and blood at week 24

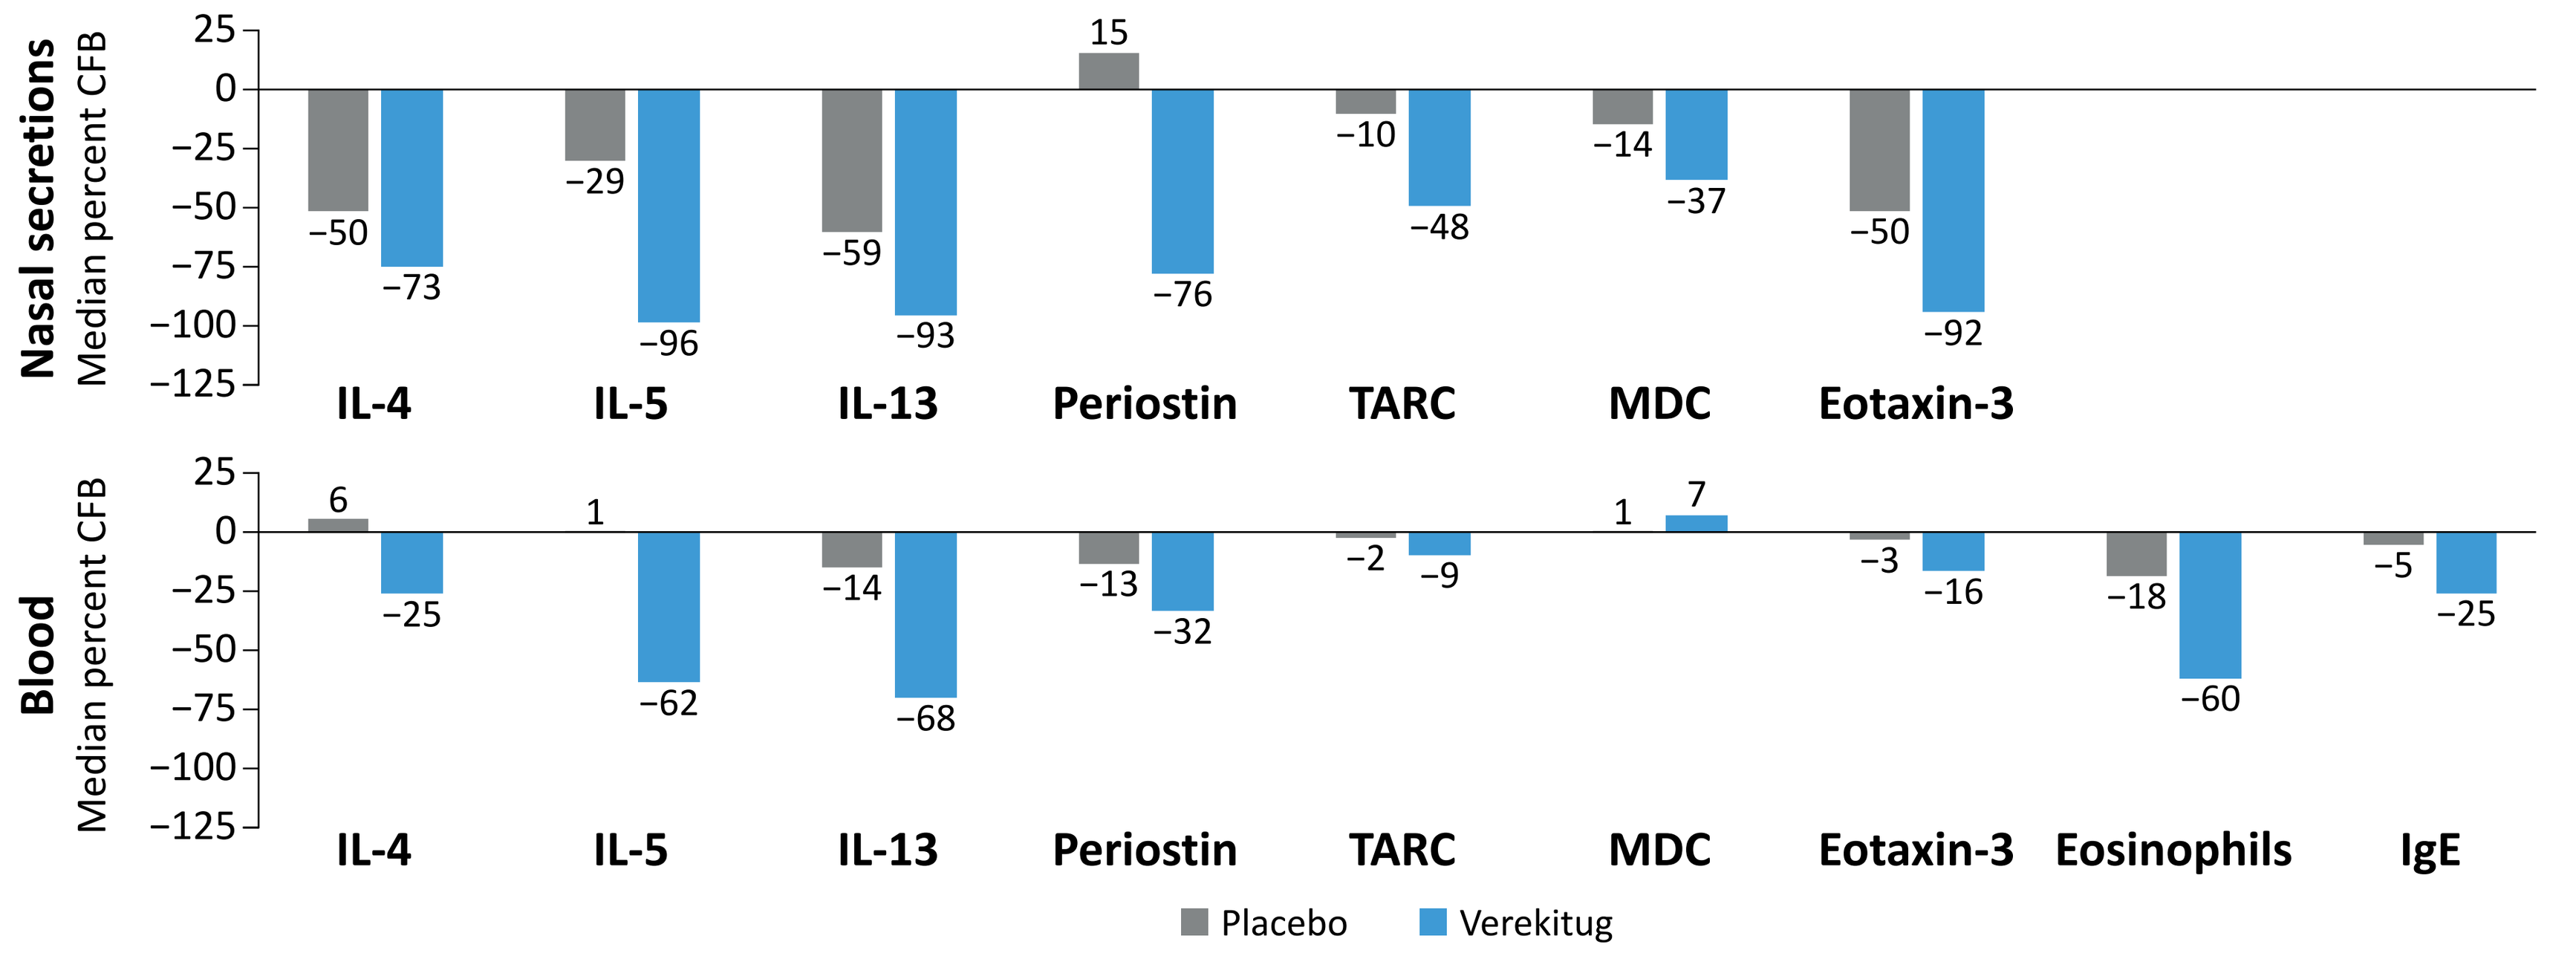

Nasal secretions
Median percent CFB
Blood
Median percent CFB
IL-4
IL-5
IL-13
Periostin
TARC
MDC
Eotaxin-3
Eosinophils
IgE
Placebo
Verekitug

IgE, immunoglobulin E; IL, interleukin; MDC, macrophage-derived chemokine; TARC, thymus and activation-regulated cytokine.

- Reductions in the cytokines **IL-4**, **IL-5**, and **IL-13**—key drivers of eosinophilic inflammation, mucus hypersecretion, and polyp growth[1]—were generally greater in nasal secretions vs in blood over 24 weeks (**Figure 3**)

Figure 3. Percent change from baseline in IL-4, IL-5, and IL-13 in nasal secretions and blood

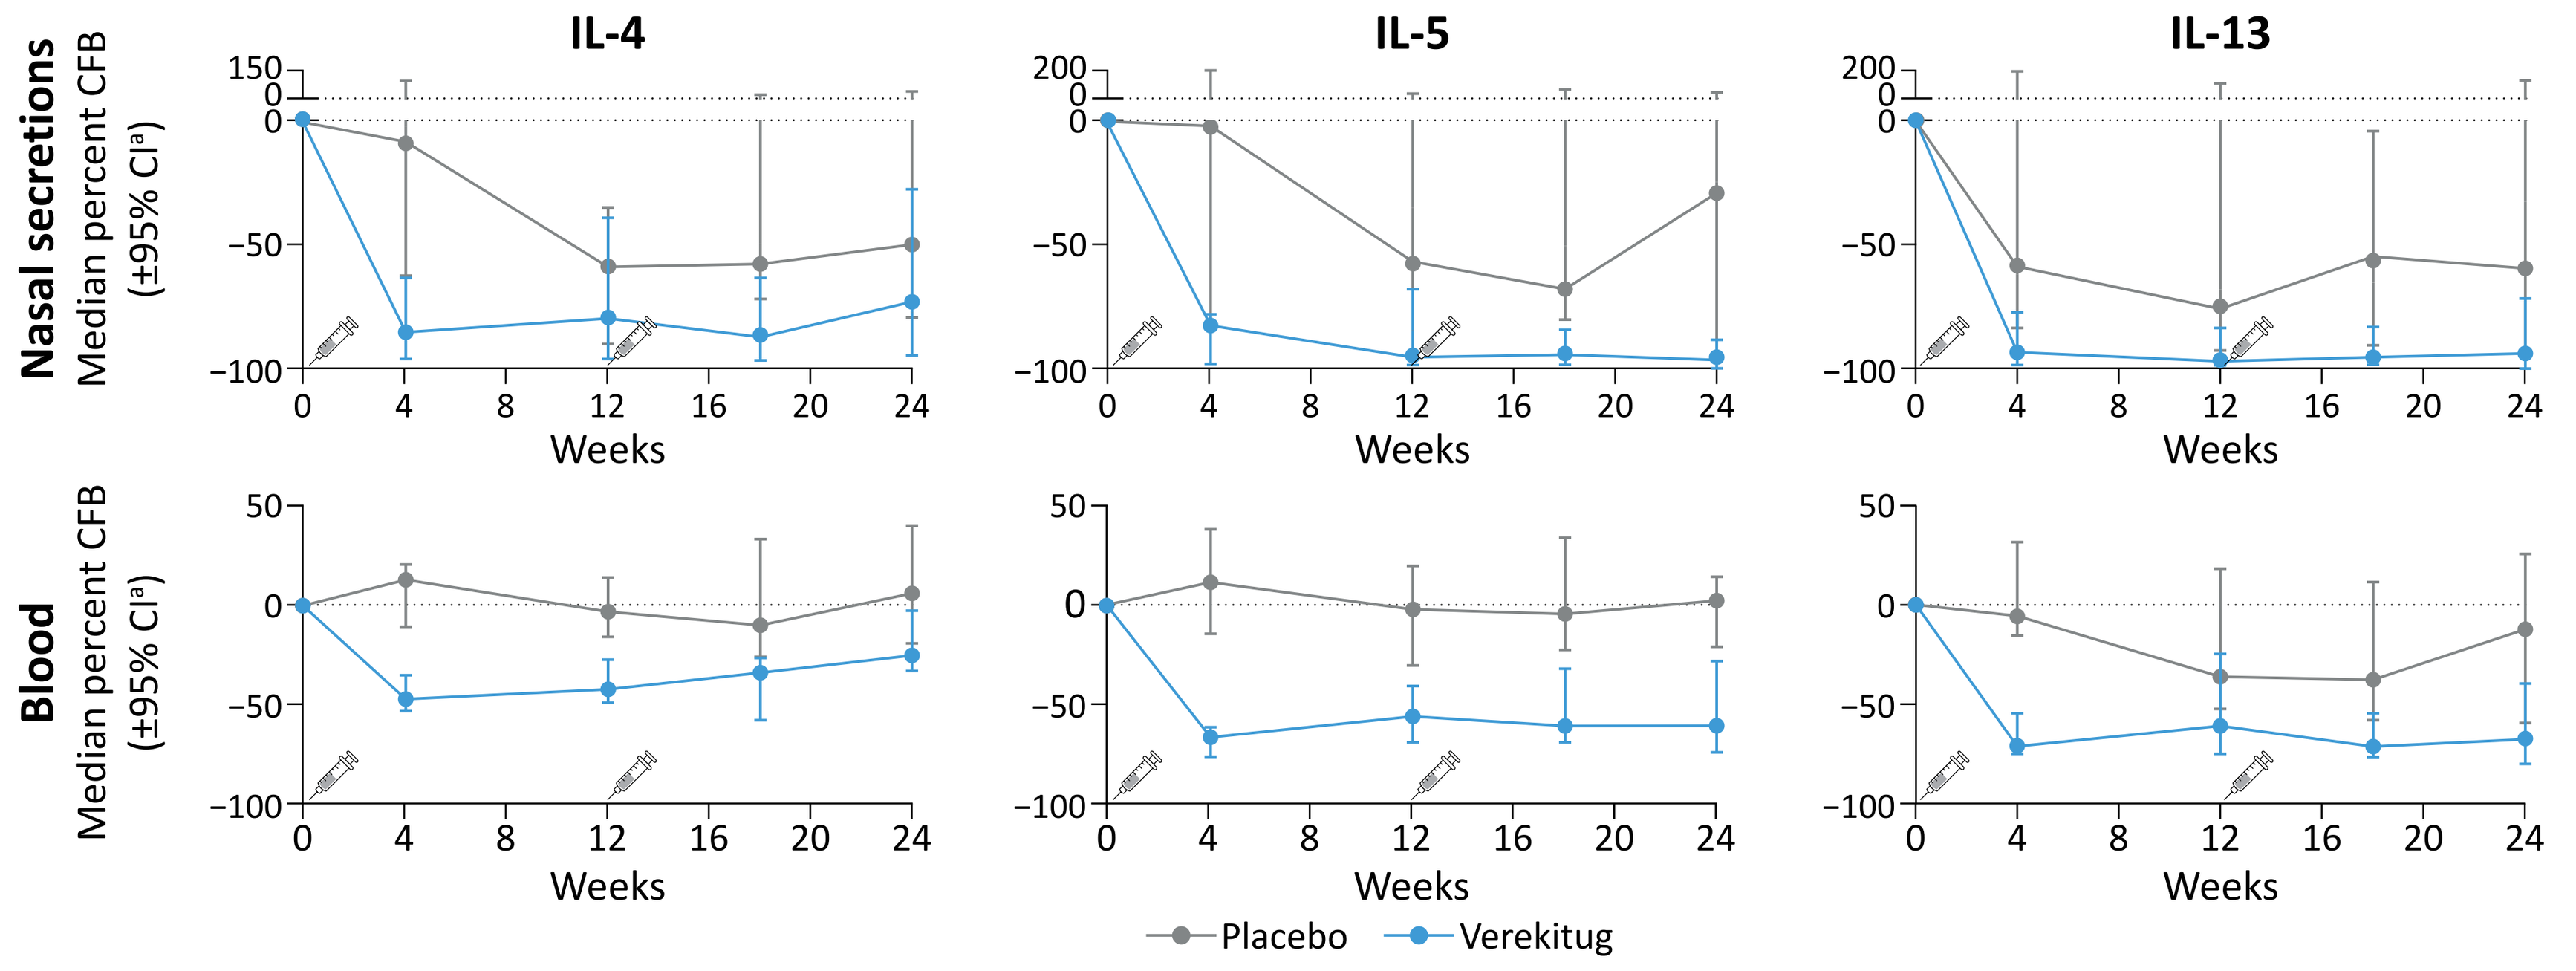

IL-4
IL-5
IL-13
Nasal secretions
Median percent CFB (±95% CI[a])
Blood
Median percent CFB (±95% CI[a])
Weeks
Placebo
Verekitug

Syringes indicate first and second dose.
[a]95% CI was calculated using GraphPad Prism version 10.6.0.

- Verekitug reduced blood **eosinophils** by 50% at week 2 and reduced **IgE** as early as week 4; reductions were sustained or ongoing over 24 weeks (**Figure 4**)

Figure 4. Percent change from baseline in blood eosinophils and IgE

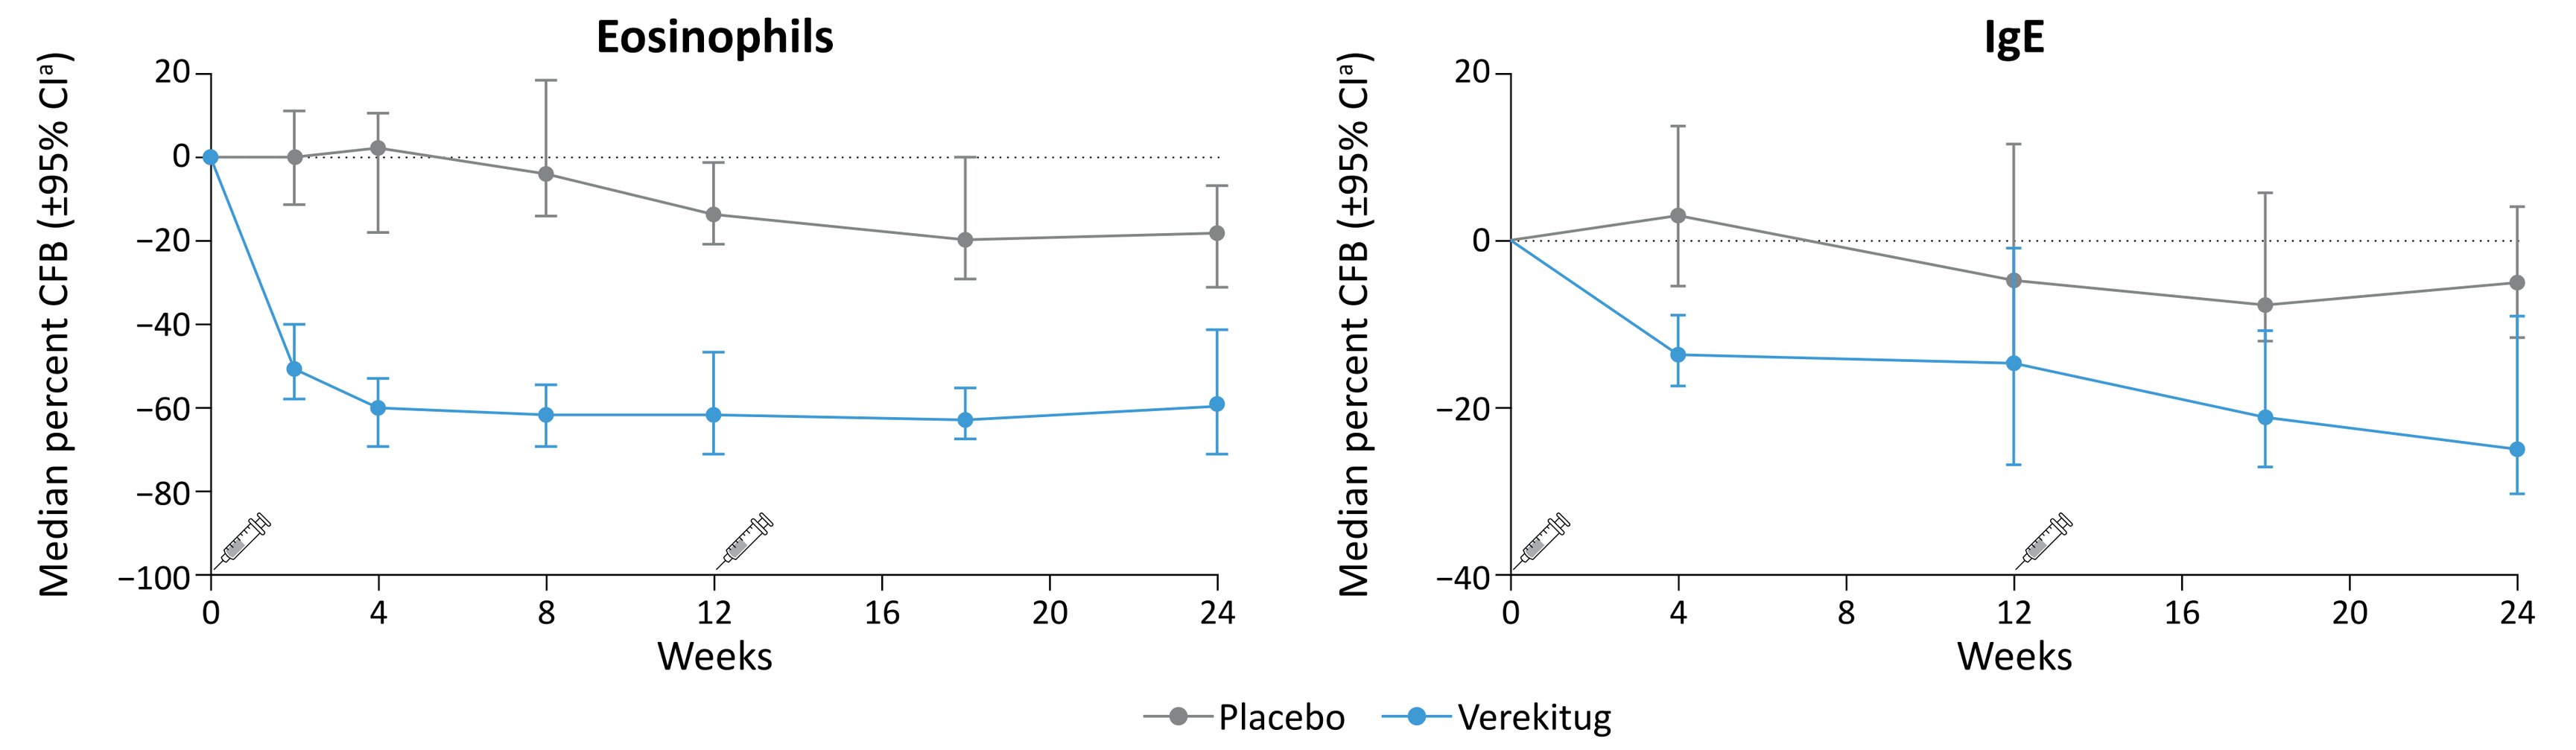

Eosinophils
IgE
Median percent CFB (±95% CI[a])
Weeks
Placebo
Verekitug

Syringes indicate first and second dose.
[a]95% CI was calculated using GraphPad Prism version 10.6.0.

- The magnitude of reduction in **IL-5**, a cytokine that plays a pivotal role in eosinophilic differentiation, activation, and survival,[1] correlated with reduction in blood **eosinophils** after verekitug treatment (**Figure 5**)

Figure 5. Correlation between reductions of IL-5 and eosinophils in blood at week 24 after verekitug treatment

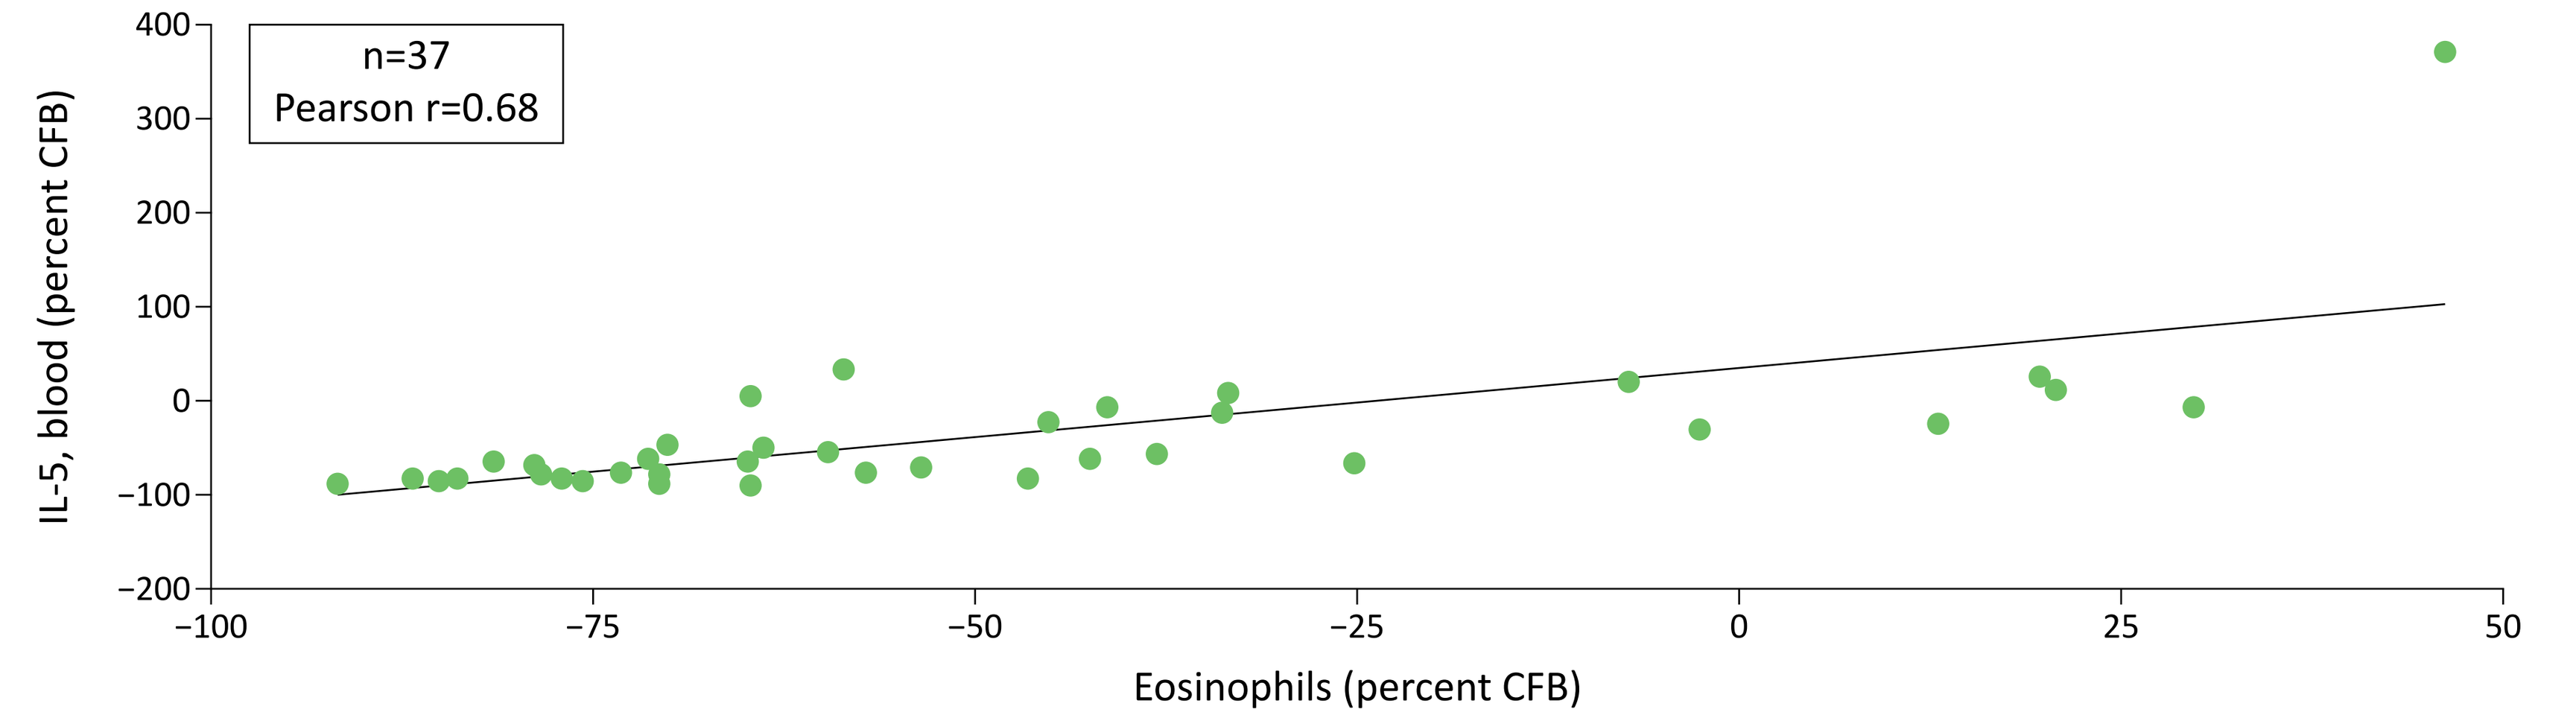

n=37
Pearson r=0.68
IL-5, blood (percent CFB)
Eosinophils (percent CFB)

CONCLUSIONS

- Verekitug 100 mg q12w led to rapid and sustained reduction of inflammatory and fibrotic biomarkers, including the canonical type 2 cytokines, IL-4, IL-5, and IL-13, in nasal secretions and blood, which may contribute to reduced disease severity, including NPS, and symptom burden
- Verekitug led to reductions in blood eosinophils, which were sustained over 24 weeks
- Verekitug led to reductions in IgE, which continued to decline over 24 weeks

Presented at the 2026 American Thoracic Society (ATS) International Conference; May 15–20; Orlando, FL, USA

P1398

Verekitug, a thymic stromal lymphopoietin receptor antagonist, in chronic rhinosinusitis with nasal polyps: effect on type 2 inflammatory biomarkers in the VIBRANT trial

Joseph K. Han,[1] Piotr H. Skarzynski,[2,3] Neetu Talreja,[4] Martha Tarpay,[5] Tanya M. Laidlaw,[6] José Miguel Villacampa Aubá,[7] Isam Alobid,[8–10] Joaquim Mullol,[8–10] Sven Becker,[11] Sumathi Sivapalasingam,[12] Kiran Patel,[12]* Ashish Kalra,[12] Arkadeep Sinha,[12] Fatema Chowdhury,[12] Richard Lin,[12] Fang Xie,[12] Andrzej Dymek,[13] Grazyna Pulka,[14] Aaron Deykin[12]

[1]Old Dominion University, Norfolk, VA, USA; [2]Center of Hearing and Speech MEDINCUS, Kajetany, Poland; [3]Institute of Sensory Organs, Kajetany, Poland; [4]The Allergy Group and Treasure Valley Medical Research, Boise, ID, USA; [5]Allergy, Asthma & Clinical Research Center, Oklahoma City, OK, USA; [6]Brigham and Women's Hospital, Harvard Medical School, Boston, MA, USA; [7]Hospital Universitario Fundación Jiménez Díaz, Madrid, Spain; [8]Hospital Clinic, Universitat de Barcelona, Barcelona, Catalonia, Spain; [9]IDIBAPS, Barcelona, Catalonia, Spain; [10]CIBERES, Barcelona, Catalonia, Spain; [11]University of Tübingen, Tübingen, Germany; [12]Upstream Bio, Inc., Waltham, MA, USA; [13]Centrum Medyczne Lucyna Andrzej Dymek, Strzelce Opolskie, Poland; [14]Centrum Medyczne ALL-MED-Badania Kliniczne, Kraków, Poland. *Affiliation at time of study.

SUPPLEMENTARY MATERIAL

Supplementary Table 1. Baseline biomarker measurements, median (range)

	Nasal secretions		Blood	
Biomarker	**Verekitug (n=29)**	**Placebo (n=31)**	**Verekitug (n=41)**	**Placebo (n=40)**
Eosinophils, cells/µL	–	–	350 (120–1230)	420 (30–2080)[a]
IgE, IU/mL	–	–	112.1 (3.5–1431)[b]	94.1 (22.6–1456)[b]
IL-4, fg/mL	94.2 (8–1613)	97.5 (8–531)	94.3 (36–420)[c]	114.5 (8–369)[c]
IL-5, fg/mL	3421 (4–295,133)	3017 (18–713,607)	1751 (398–37,413)[c]	1814 (683–72,415)[c]
IL-13, fg/mL	442.0 (16–31,758)	233.0 (16–33,020)	47.5 (16–447)[c]	46.9 (16–418)[c]
Periostin, ng/mL	64.1 (0.6–1162.5)	59.1 (0.2–812.1)	32.3 (14.2–73.8)[c]	33.2 (17.8–78.4)[c]
TARC, pg/mL	8.9 (1–75)	4.6 (1–19)	509.0 (103–1602)[c]	412.5 (131–1688)[c]
MDC, pg/mL	15.6 (2–139)	16.7 (2–111)	959.5 (146–2101)[c]	853.0 (509–2735)[c]
Eotaxin-3 levels, pg/mL	24.7 (2–1493)	15.5 (2–464)	29.3 (10–5511)[c]	27.0 (2–362)[c]
IFN-ϒ levels, fg/mL	5.6 (6–2469)	29.3 (6–3298)	502.5 (138–1753)[c]	453.5 (160–33,665)[c]
TNF-α levels, fg/mL	158 (13–13,733)	340 (22–7133)	787 (372–2053)[c]	744 (453–1724)[c]
IL-17A levels, fg/mL	459 (29–41,633)	651 (29–4245)	474 (143–3621)[c]	589 (136–2040)[c]
TSLP, fg/mL	1605 (224–34,547)	2755 (248–49,552)[d]	1205 (681–3449)[c]	1274 (464–4005)[c]

[a]n=39. [b]n=33. [c]n=38. [d]n=30.
IgE, immunoglobulin E; IFN, interferon; IL, interleukin; MDC, macrophage-derived chemokine; TARC, thymus and activation-regulated cytokine; TNF, tumor necrosis factor; TSLP, thymic stromal lymphopoietin.

Presented at the 2026 American Thoracic Society (ATS) International Conference; May 15–20; Orlando, FL, USA